Novel NLRP3 Inflammasome Inhibitor OLT1177 Reduces Infarct Size in a Mouse Model of Myocardial Ischemia Reperfusion Injury

VCU Health
Pauley Heart Center
School of Pharmacy
Victoria Johnson Research Laboratory

Stefano Toldo[1], Adolfo G. Mauro[1], Carlo Marchetti[2], Eleonora Mezzaroma[1], Benjamin Van Tassell[1], Damaris Skouras[3], Charles Dinarello[2], Antonio Abbate[1]

1 Virginia Commonwealth University, 2 University of Colorado, 3 Olatec Therapeutics LLC.

Background

Activation of the NLRP3 inflammasome during acute myocardial infarction (AMI) mediates further injury after reperfusion.

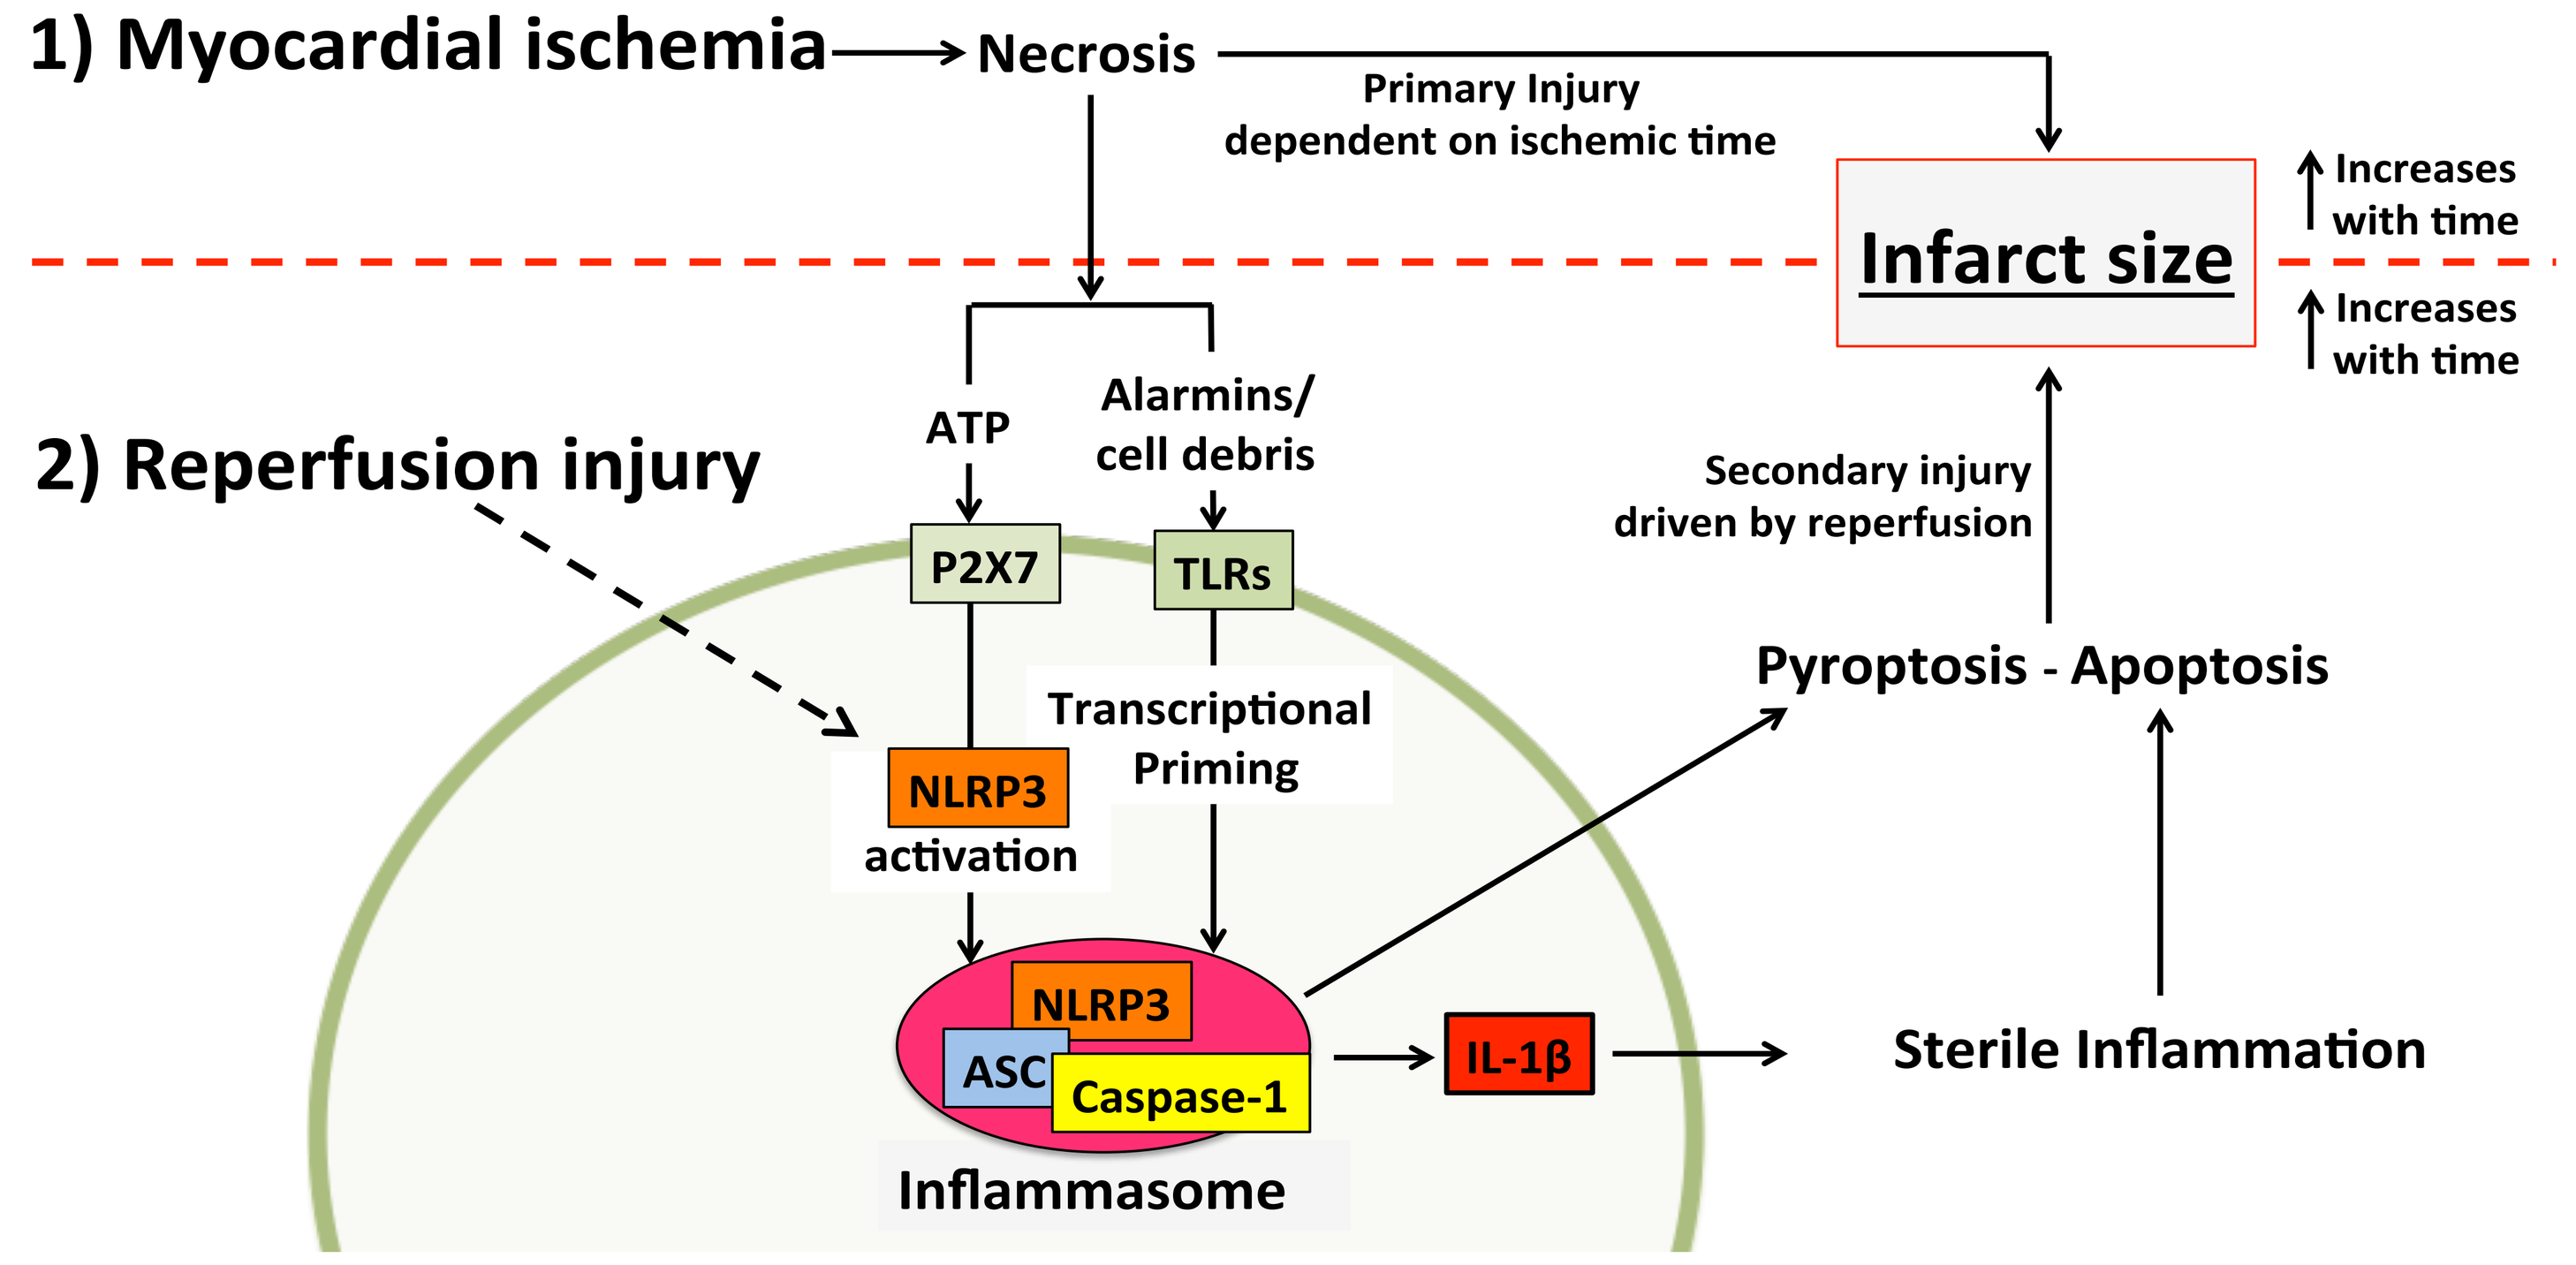
1) Myocardial ischemia
Necrosis
Primary Injury
dependent on ischemic time
Infarct size
Increases with time
Increases with time
2) Reperfusion injury
ATP
Alarmins/ cell debris
P2X7
TLRs
Secondary injury driven by reperfusion
Pyroptosis - Apoptosis
Transcriptional Priming
NLRP3 activation
NLRP3
ASC
Caspase-1
IL-1β
Sterile Inflammation
Inflammasome

NLRP3 inflammasome inhibitors have been shown to reduce ischemia/reperfusion injury in preclinical models, yet effective and safe therapies are still not available for patients with AMI.

OLT1177, is a novel, β-sulfonyl nitrile synthetic small molecule NLRP3 inflammasome inhibitor, with a favorable Phase 1 safety and tolerability profile in healthy volunteers, and is currently being studied in a Phase 2 clinical trial in acute gout.

Aim of the Study

To determine the efficacy of OLT1177 in an experimental mouse AMI model.

Methods

Male CD-1 mice underwent transient surgical ligation of the left coronary artery for 30 minutes followed by reperfusion for 24 hours. We administered OLT1177 (6, 60, or 600 mg/kg) or matching vehicle intraperitoneally at time of reperfusion or OLT1177 (60 mg/kg) after a 60-, 120- or 180-minute delay (N=4-8/group). Echocardiography was performed to measure left ventricular (LV) fractional shortening (FS). Infarct size was measured at pathology by triphenyl-tetrazolium-chloride (TTC) and phthalo blue staining and expressed as a percentage of the myocardium at risk.

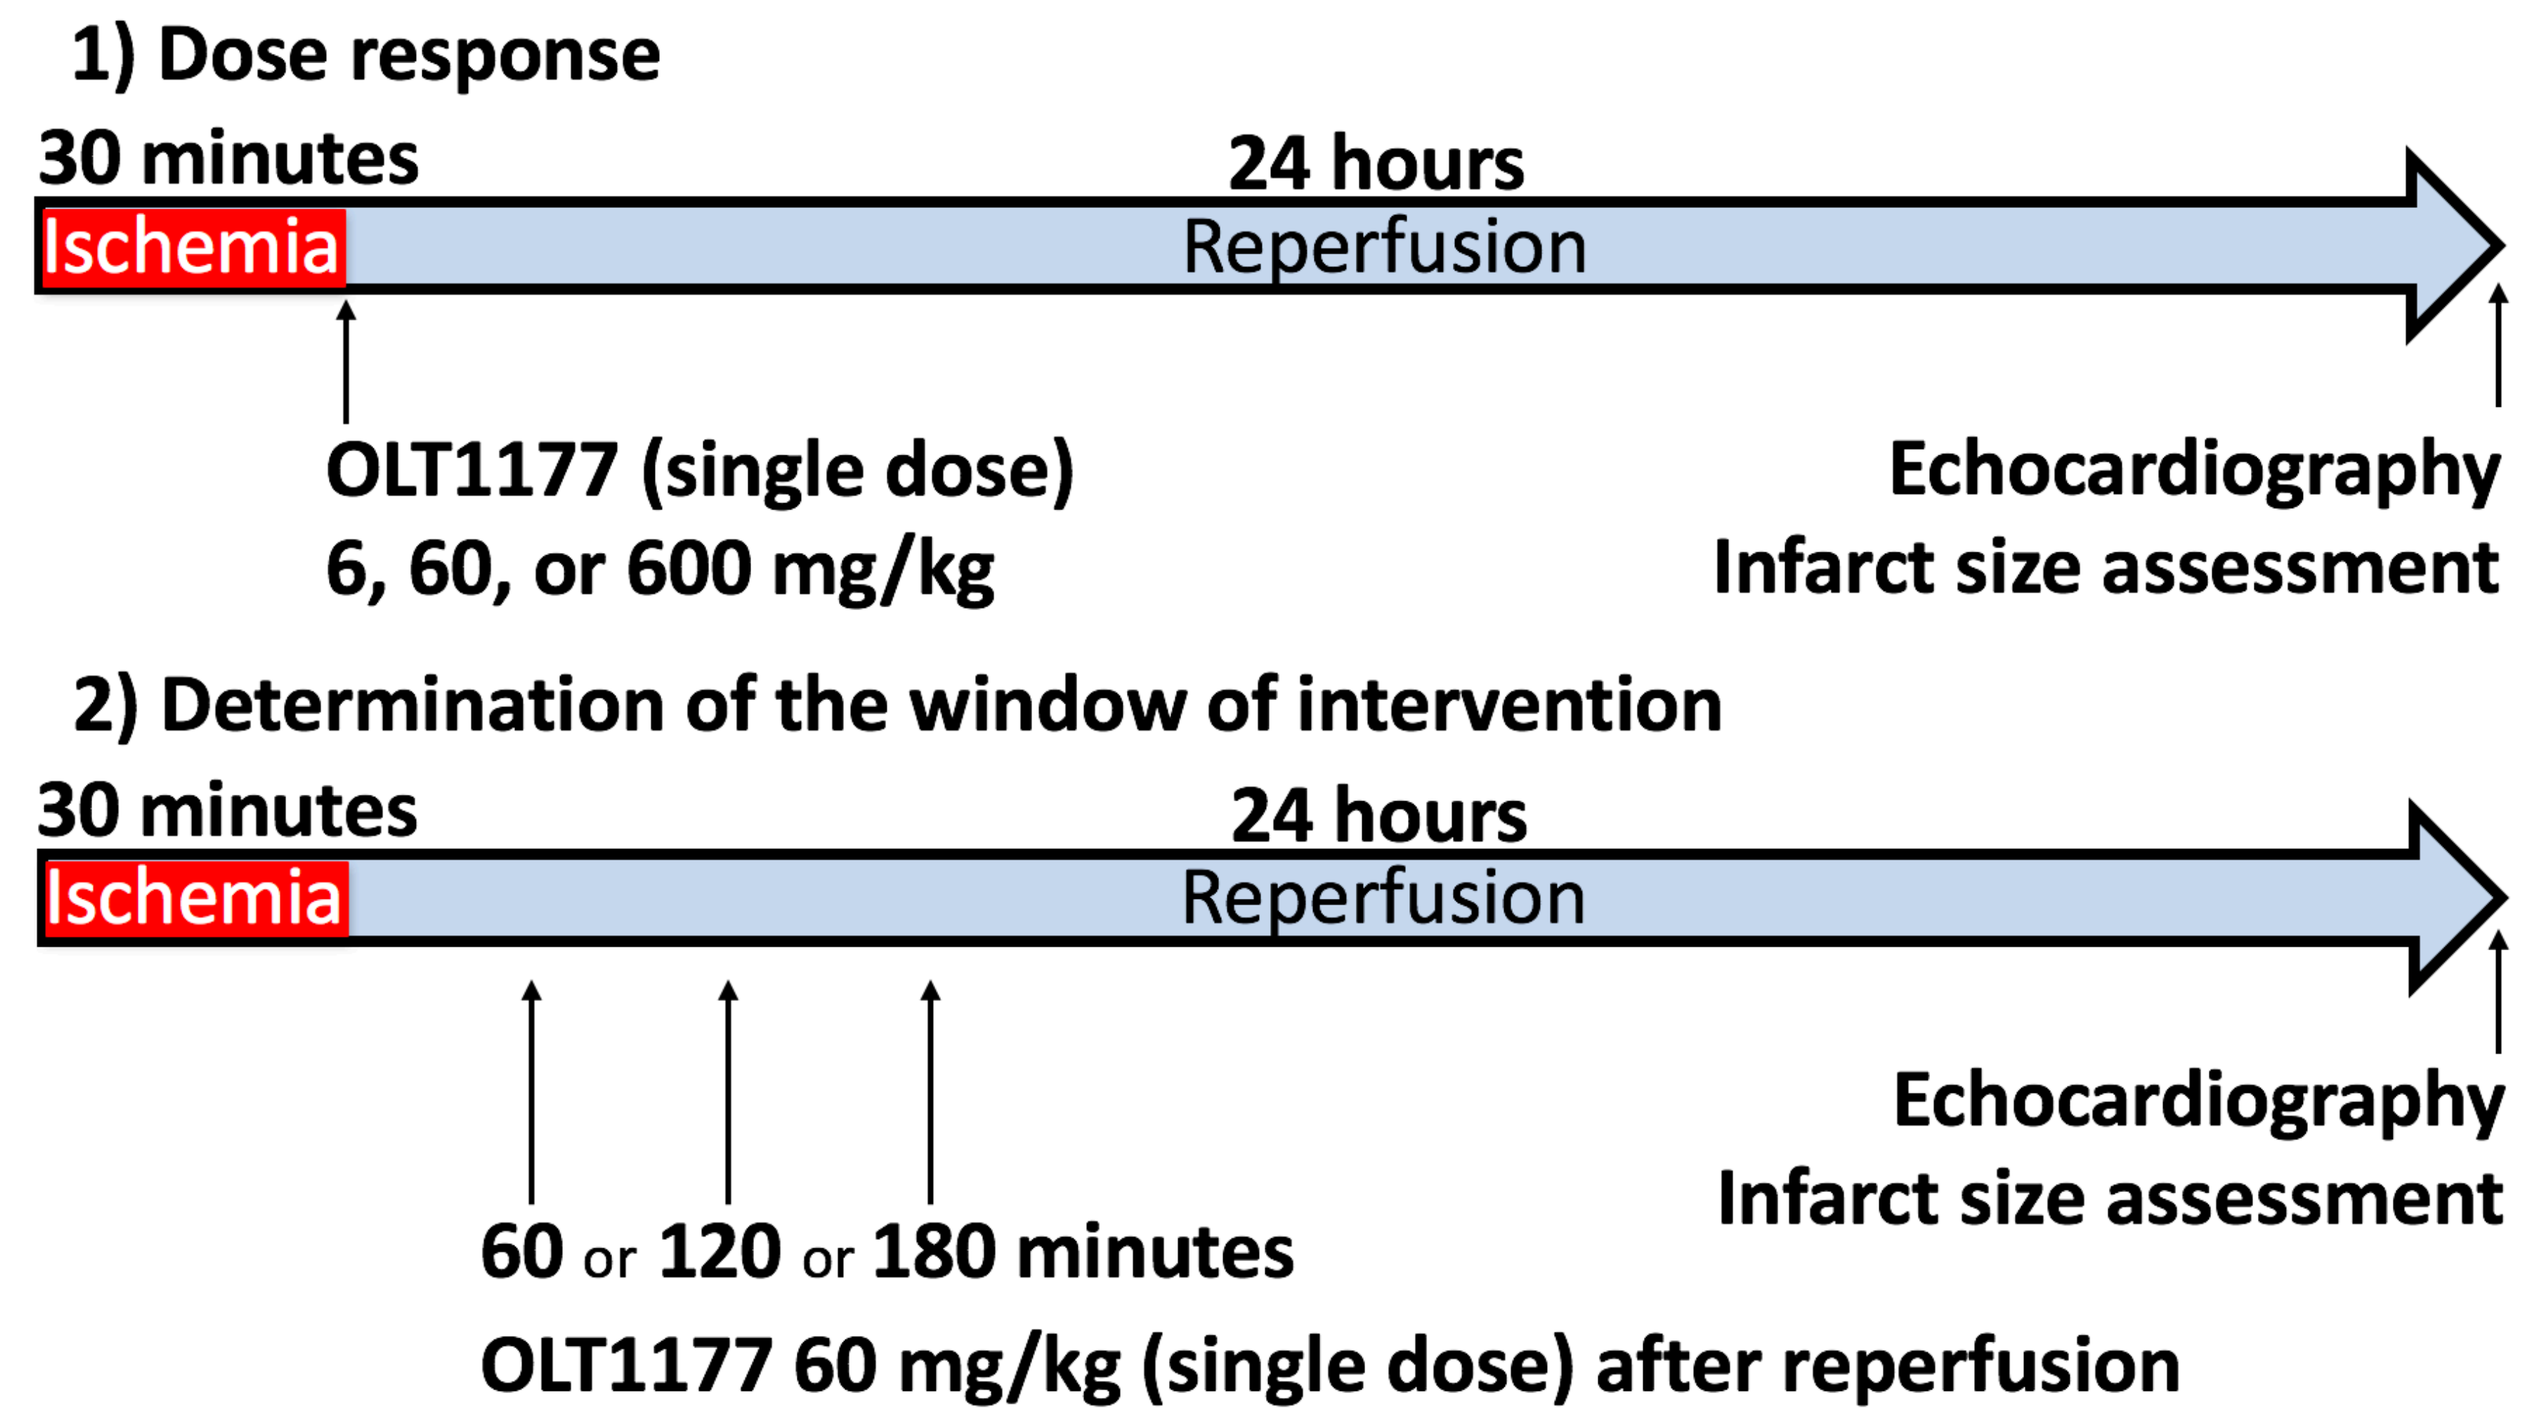
1) Dose response
30 minutes
24 hours
Ischemia
Reperfusion
OLT1177 (single dose)
6, 60, or 600 mg/kg
Echocardiography
Infarct size assessment
2) Determination of the window of intervention
30 minutes
24 hours
Ischemia
Reperfusion
Echocardiography
Infarct size assessment
60 or 120 or 180 minutes
OLT1177 60 mg/kg (single dose) after reperfusion

Model Validation

Myocardial NLPR3 expression and caspase-1 activation increase between 3-6 hours following reperfusion and peak at 24 hours.

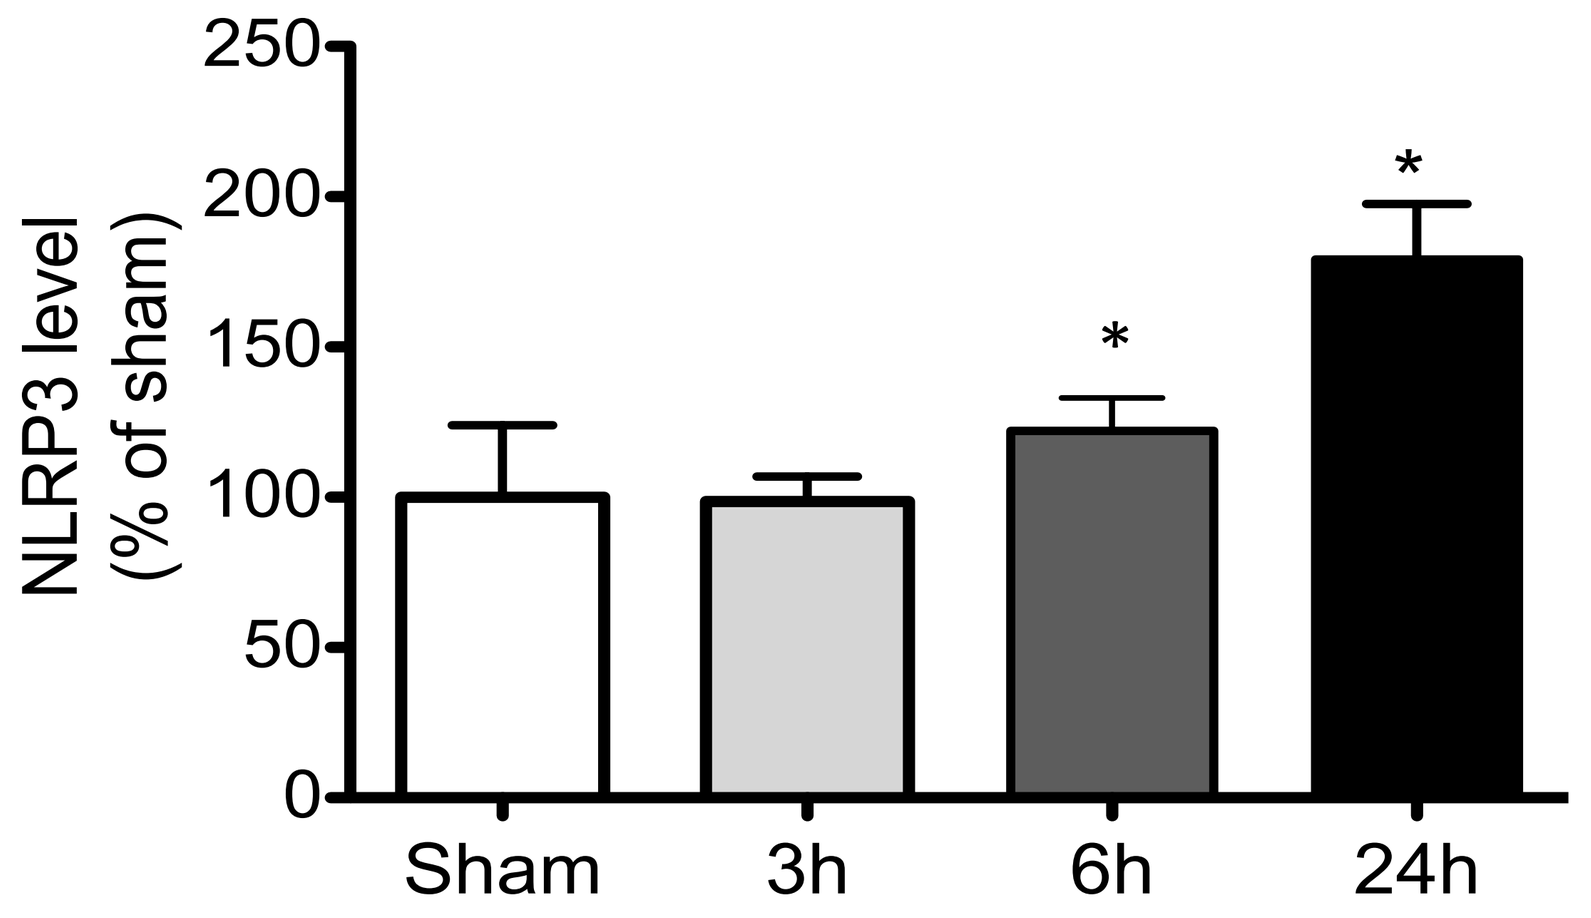
NLRP3 level (% of sham)
250
200
150
100
50
0
*
*
Sham
3h
6h
24h

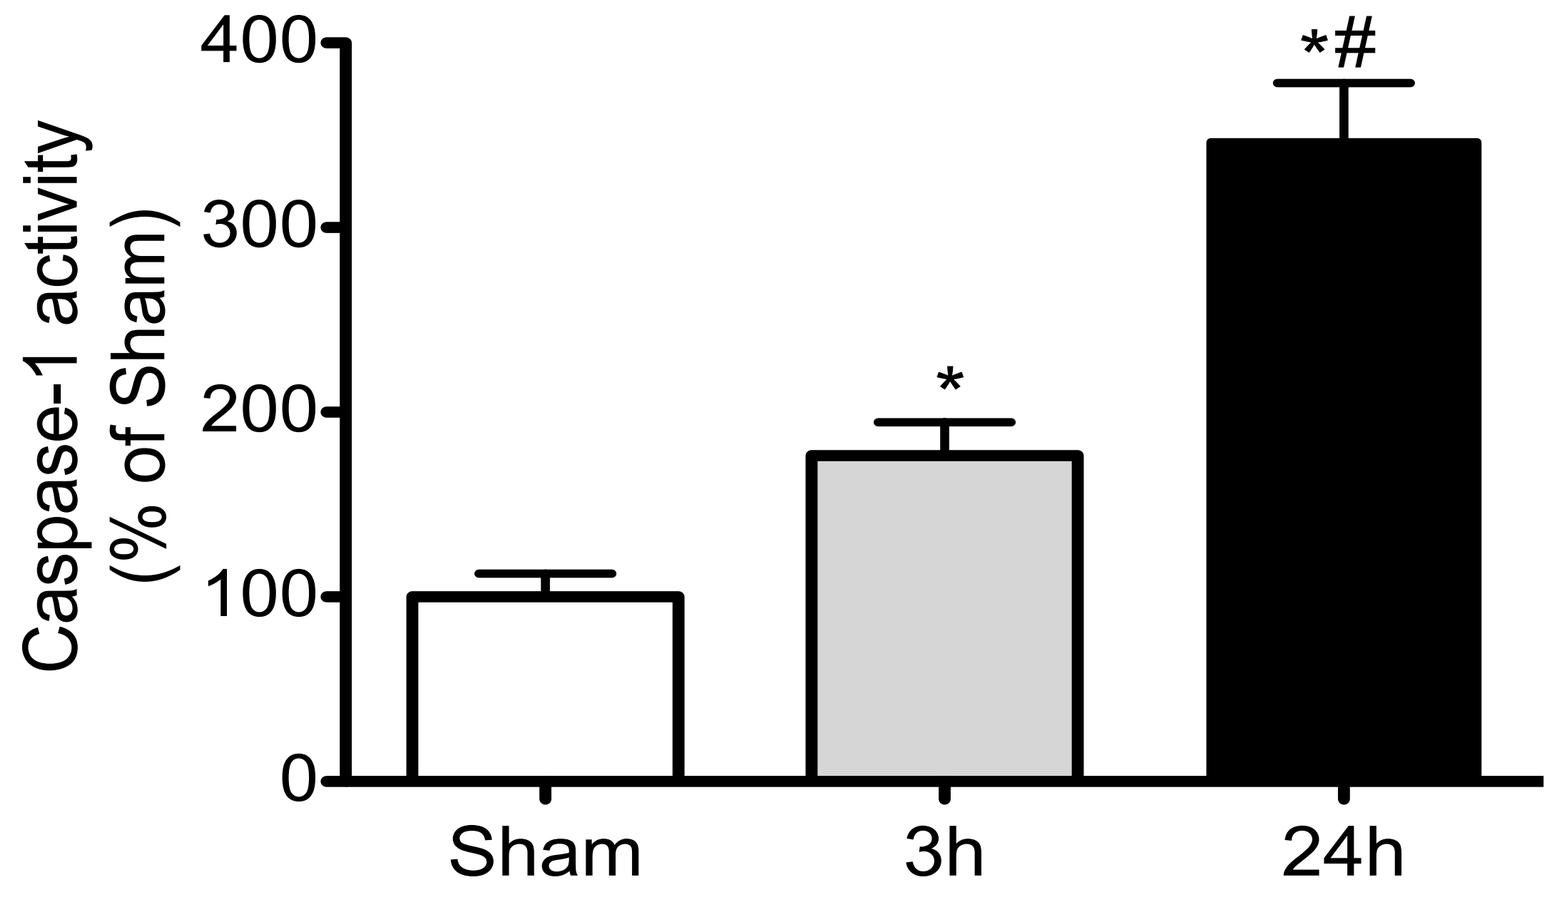
Caspase-1 activity (% of Sham)
400
300
200
100
0
*
*#
Sham
3h
24h

The left panel shows an increased expression of NLRP3 protein in the myocardium at 6 and 24 hours following reperfusion compared to 3 hours after ischemia-reperfusion injury and the sham surgery. The right panel shows a significant increase in caspase-1 activity, measured using a fluorometric enzymatic assay, reflecting inflammasome activation in the heart, at 24 hours and 3 hours versus sham. *P<0.05 vs Sham. #P<0.05 vs 3h. Extracted from Toldo S. et al. Int J Cardiol. 2016.

Results

OLT1177 (60 mg/kg) given at reperfusion provided a significant reduction in infarct size associated with a preservation of left ventricular systolic function.

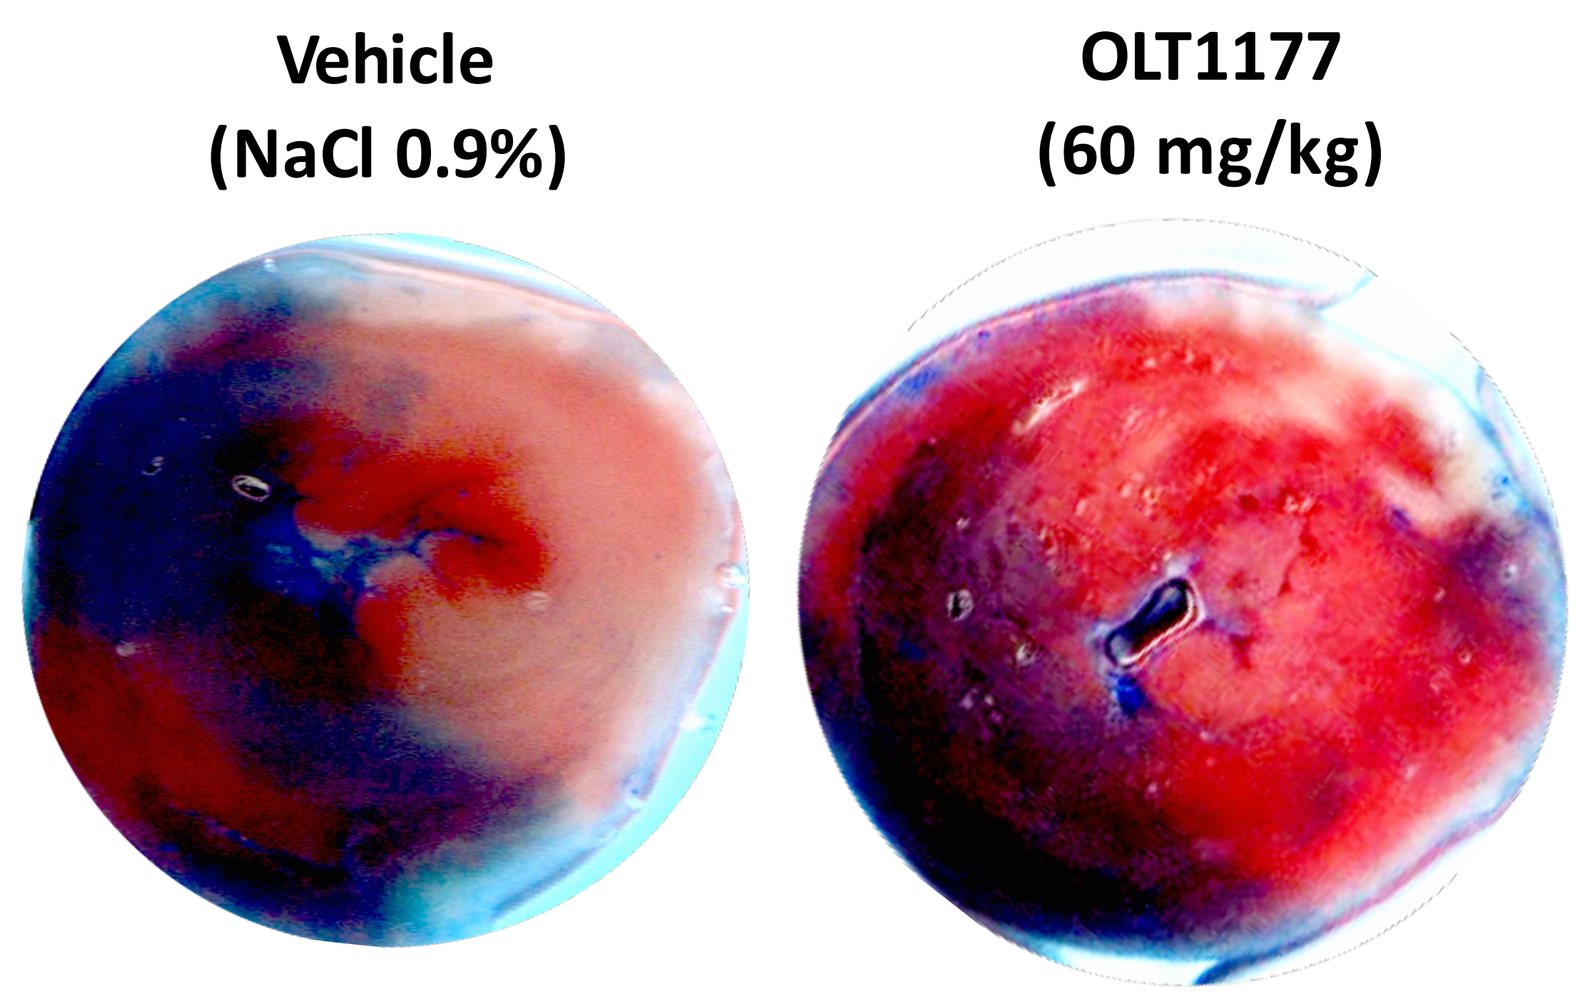
Vehicle
(NaCl 0.9%)
OLT1177
(60 mg/kg)

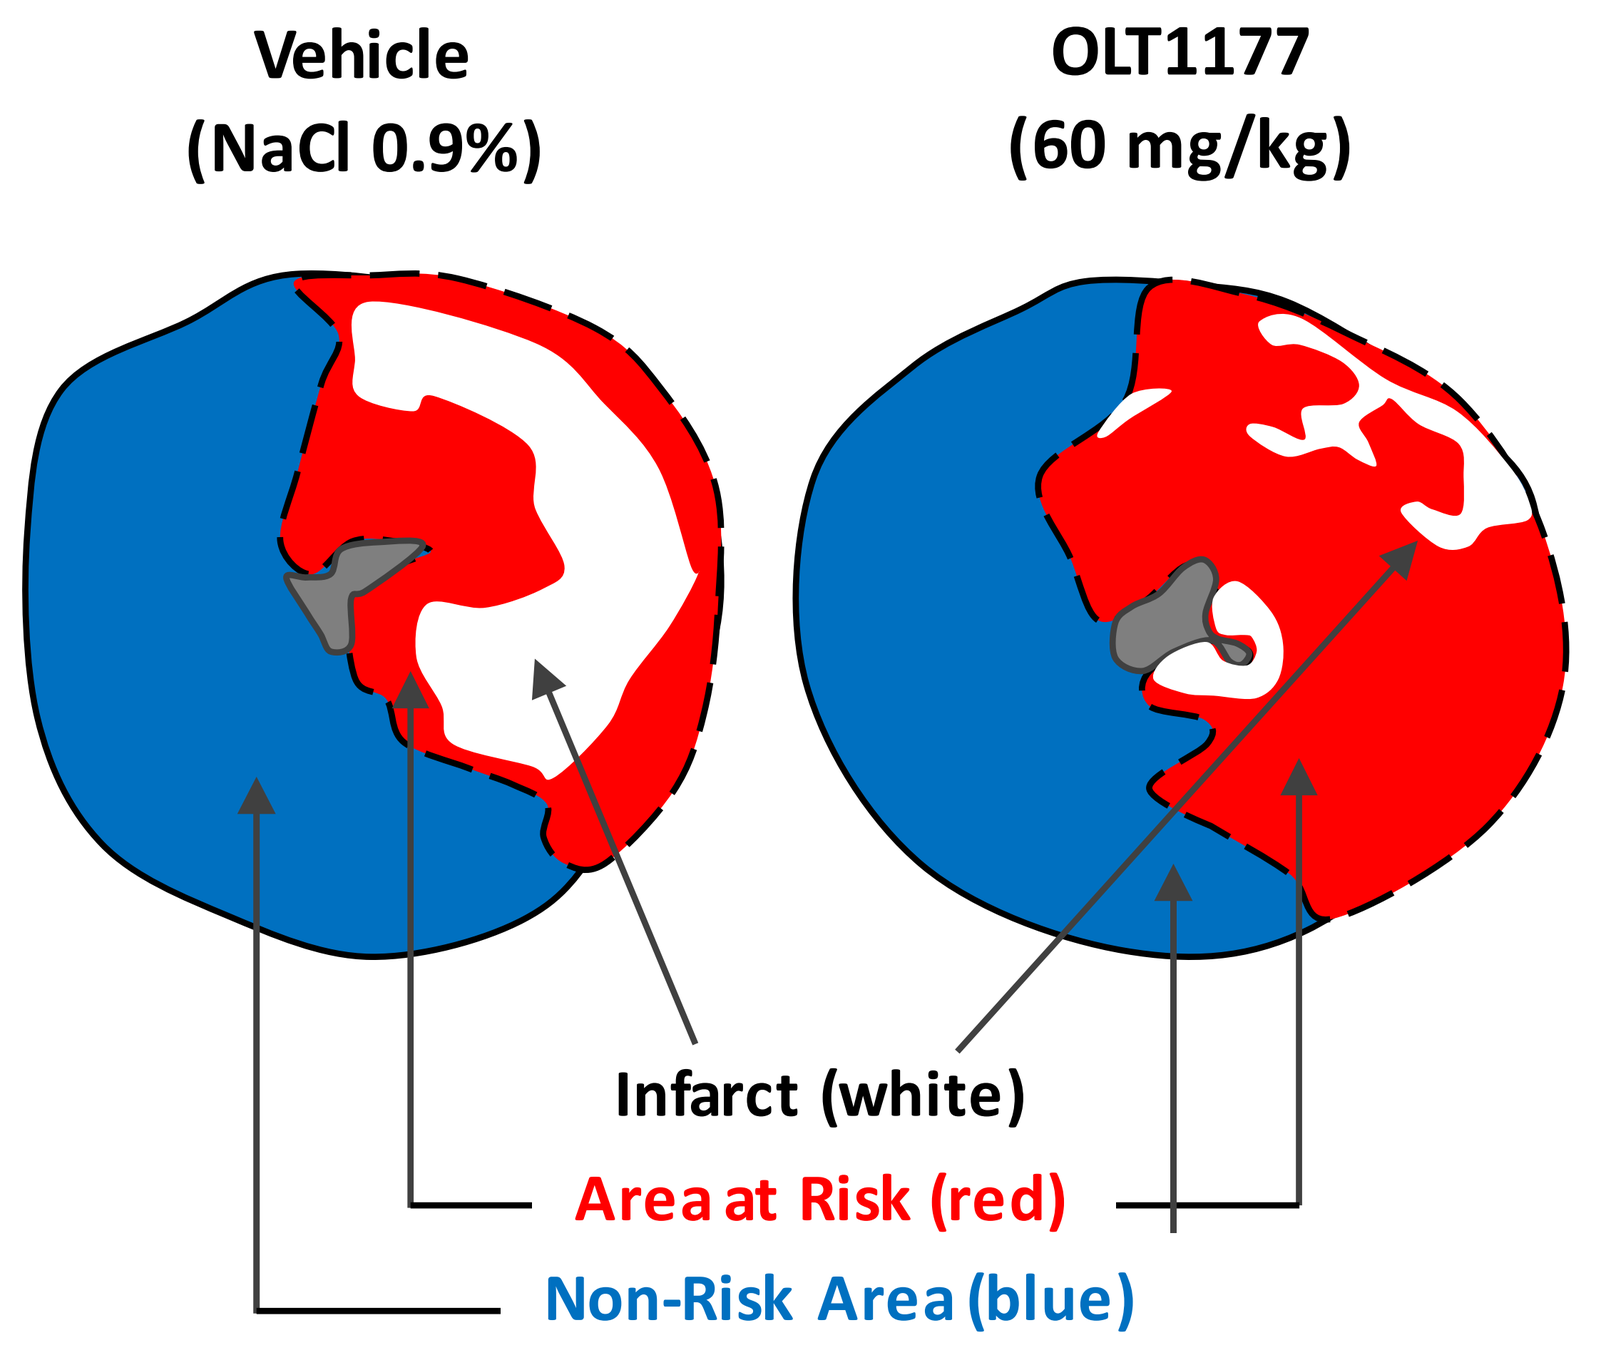
Vehicle
(NaCl 0.9%)
OLT1177
(60 mg/kg)
Infarct (white)
Area at Risk (red)
Non-Risk Area (blue)

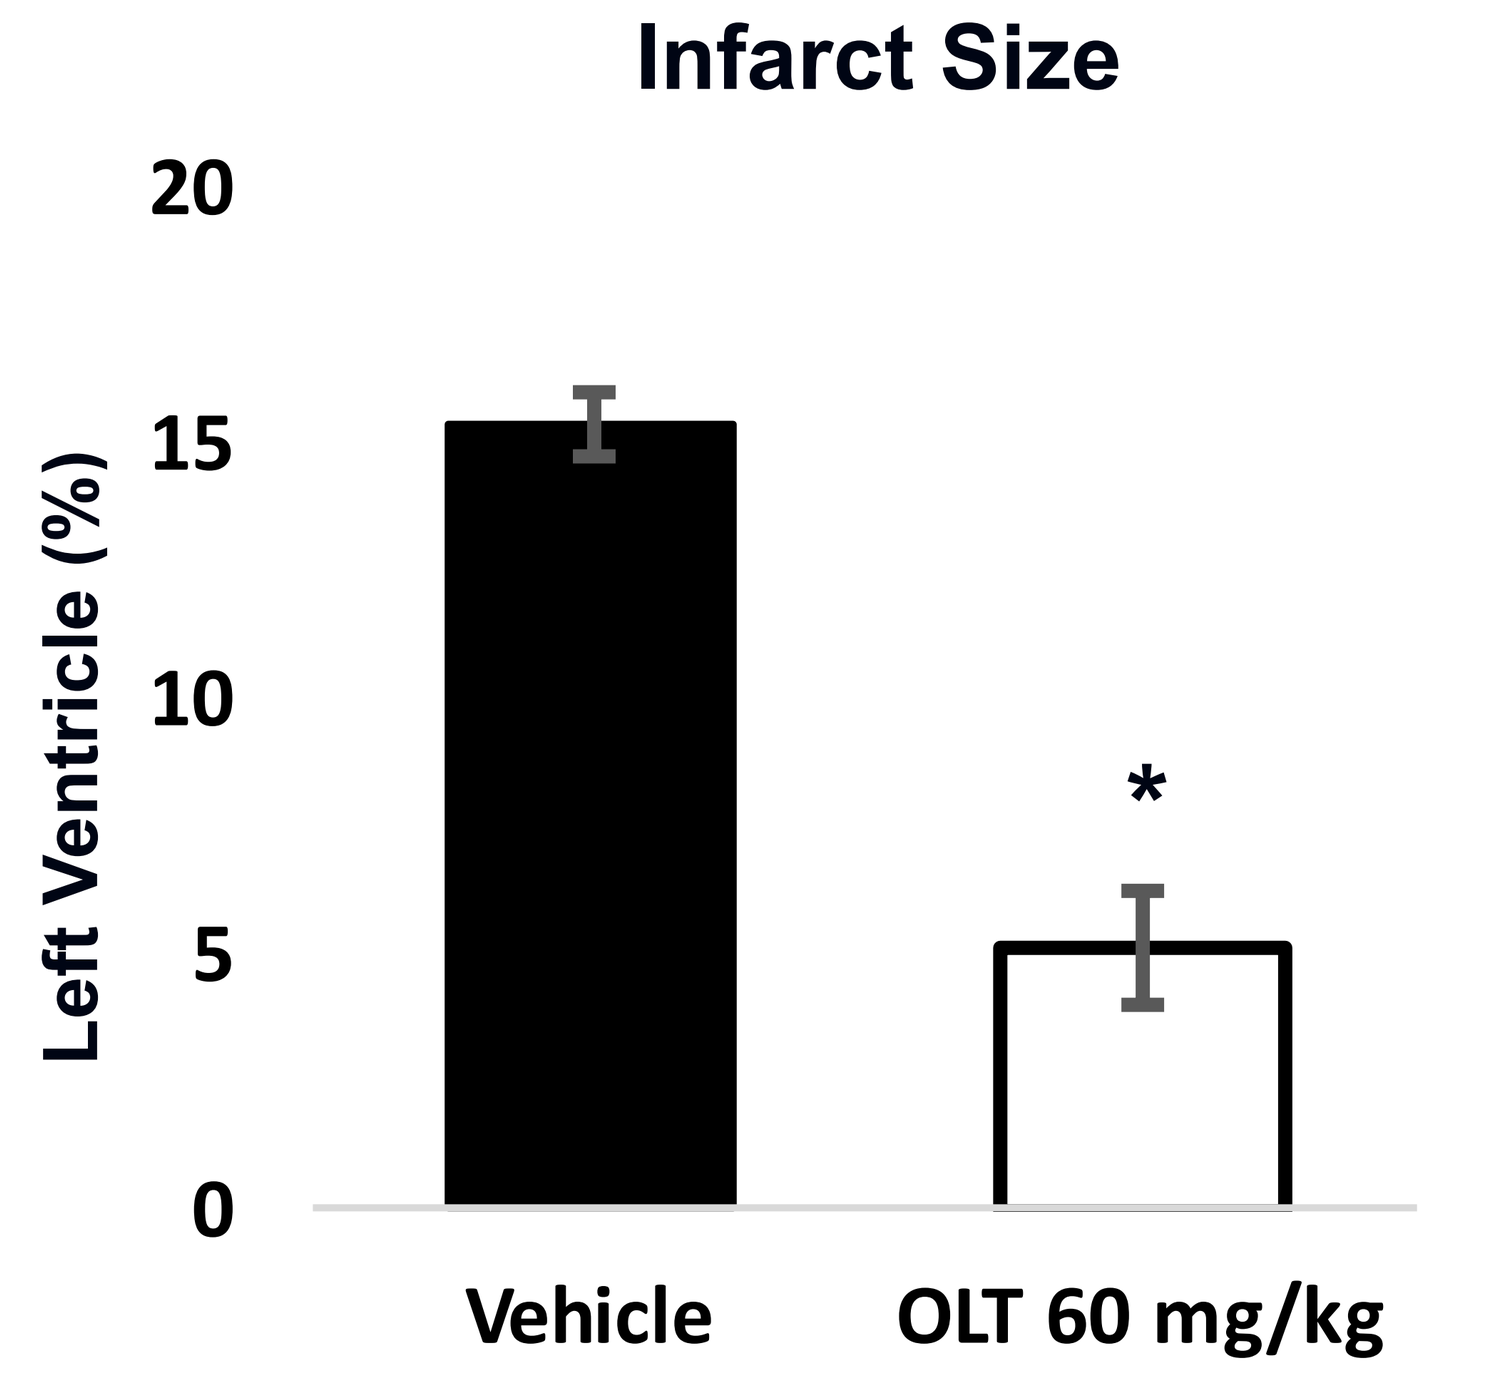
Infarct Size
Left Ventricle (%)
20
15
10
5
0
*
Vehicle
OLT 60 mg/kg

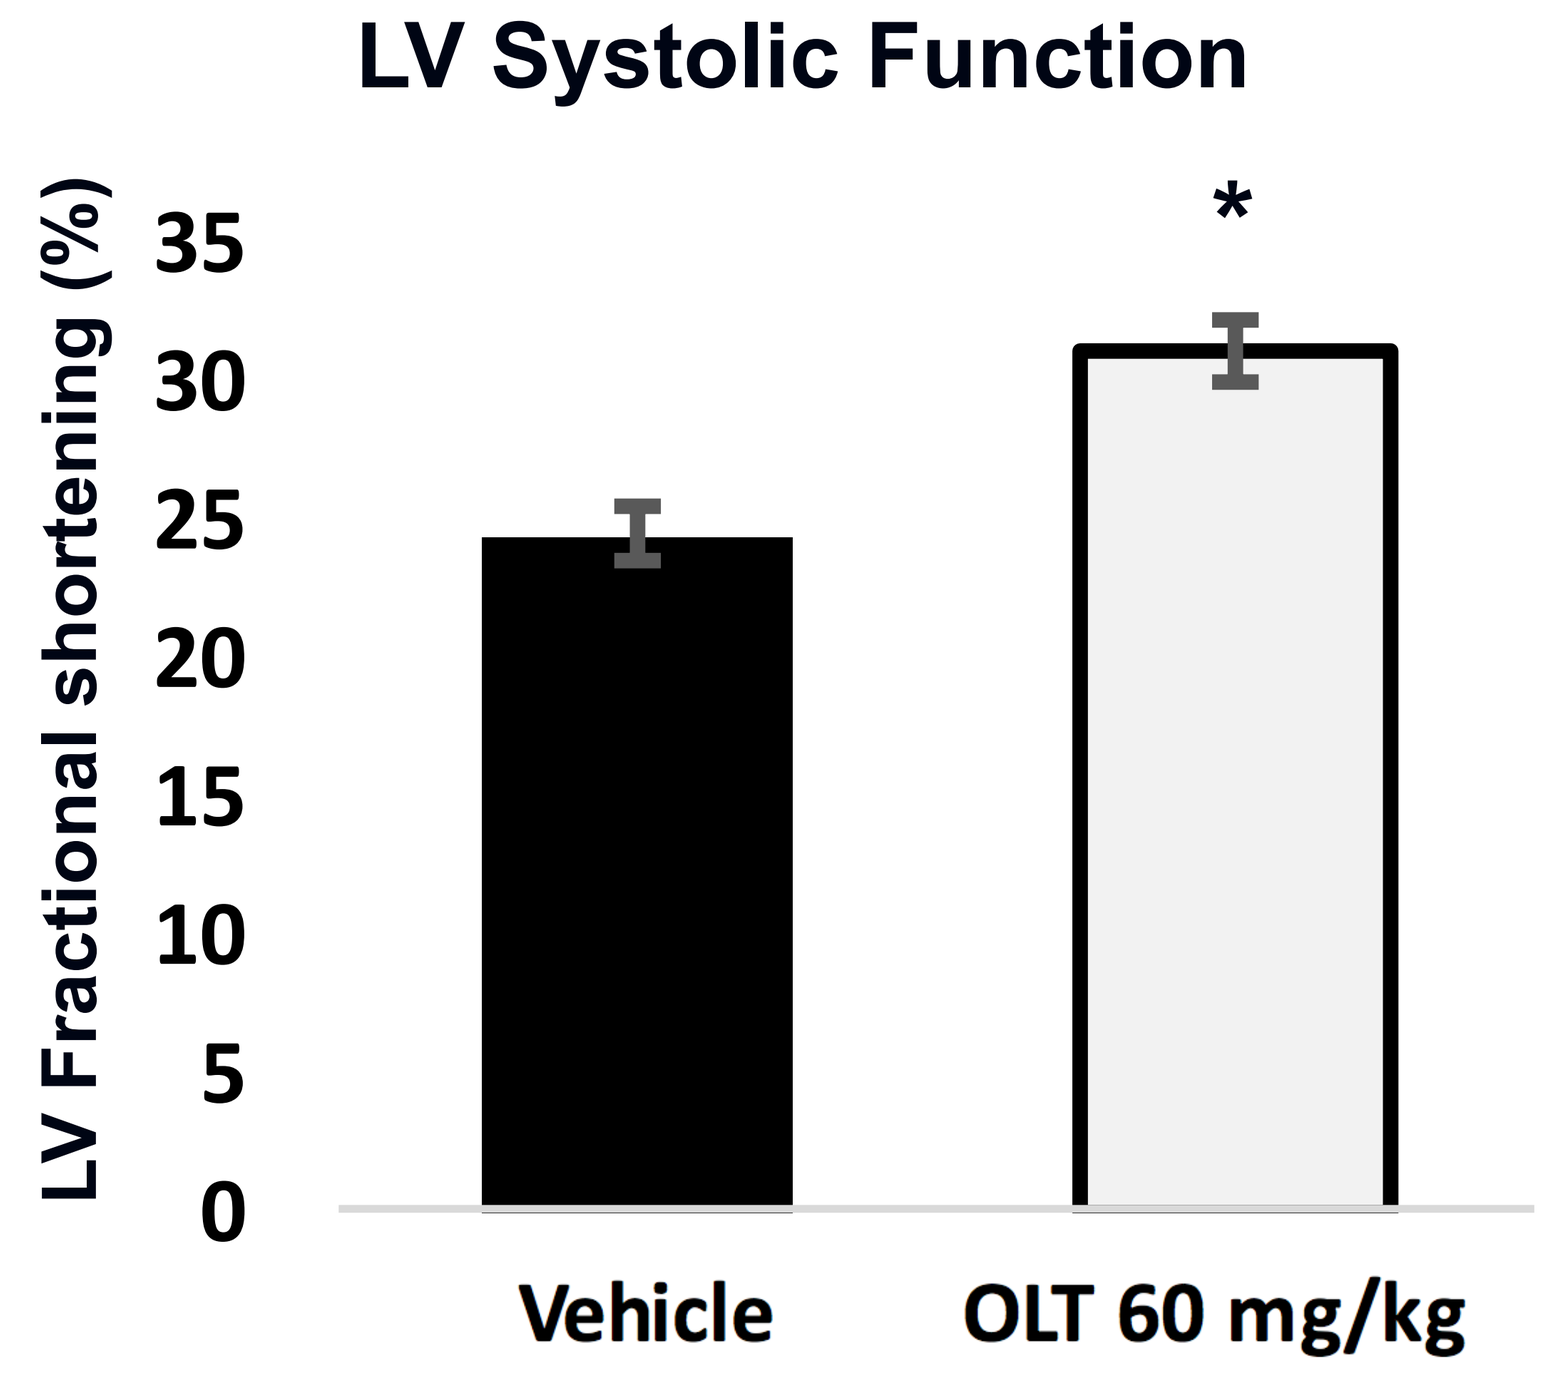
LV Systolic Function
LV Fractional shortening (%)
35
30
25
20
15
10
5
0
*
Vehicle
OLT 60 mg/kg

Effects of OLT1177 on infarct size and LV systolic function. OLT1177 (60 mg/kg) was given at the time of reperfusion. N=6-8 per group. * P<0.005

We observed no differences in infarct size between doses of 60 and 600 mg/kg. The effect on infarct size with 6 mg/kg was milder when compared to the 60 mg/kg dose. Fractional shortening was equally preserved at 24 hours with all doses of OLT1177.

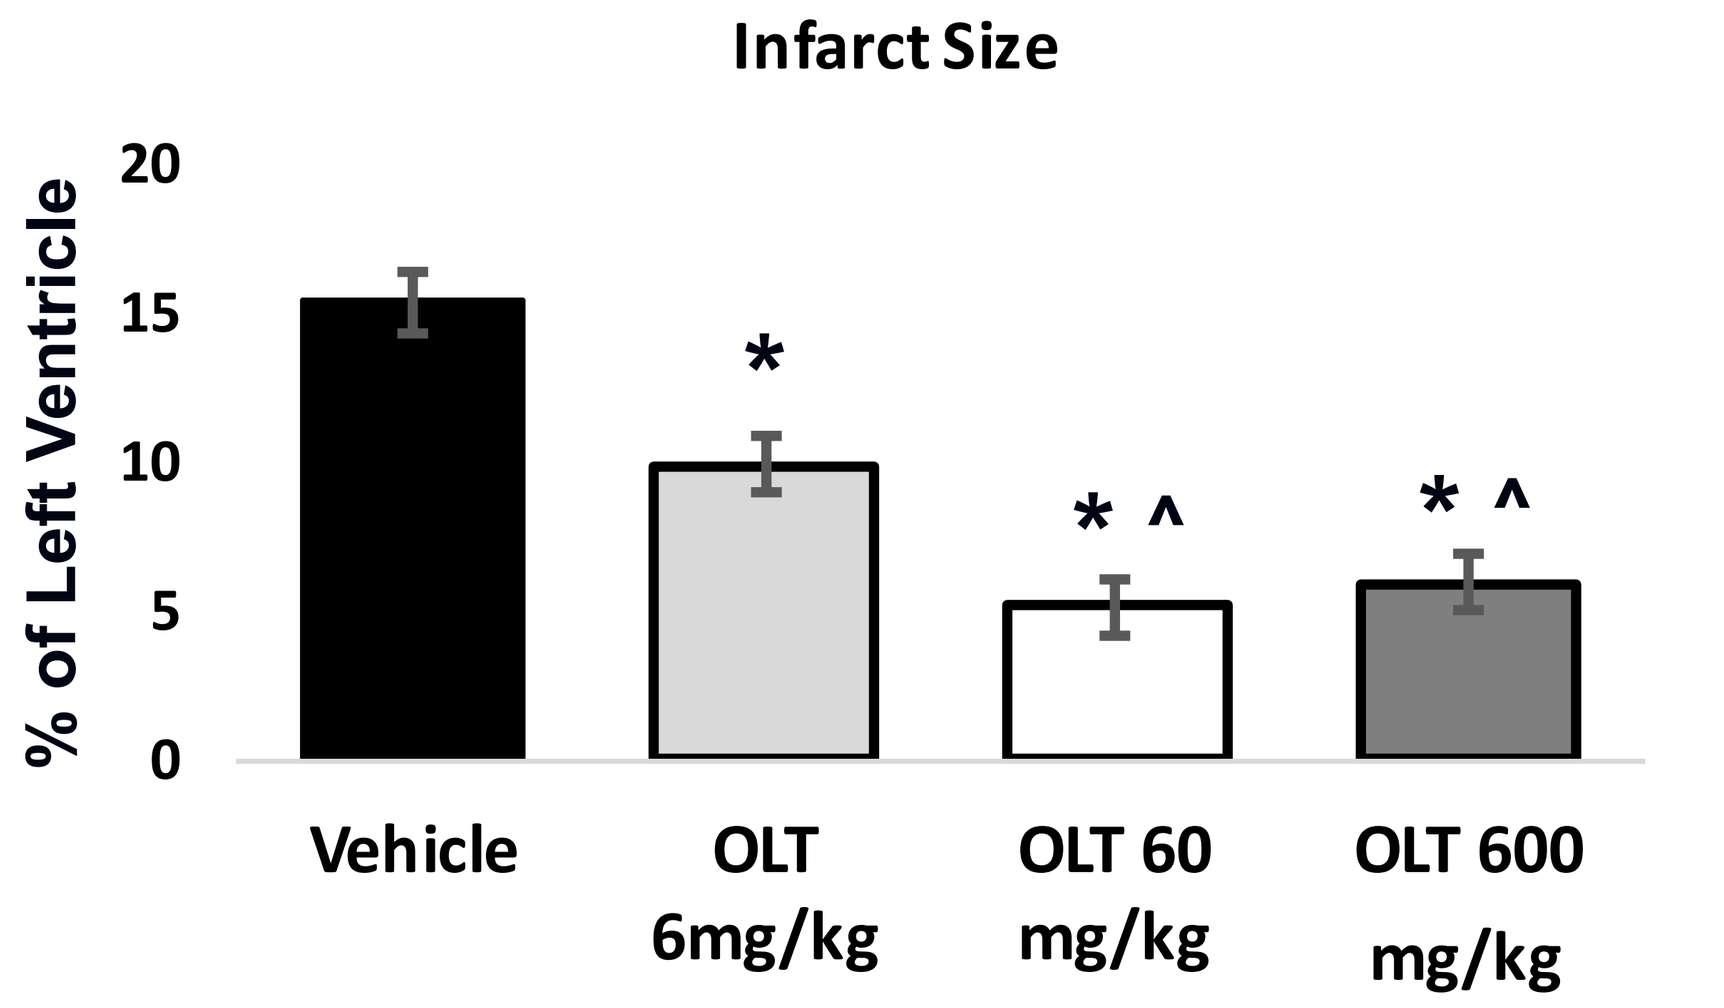
Infarct Size
% of Left Ventricle
20
15
10
5
0
*
* ^
* ^
Vehicle
OLT 6mg/kg
OLT 60 mg/kg
OLT 600 mg/kg

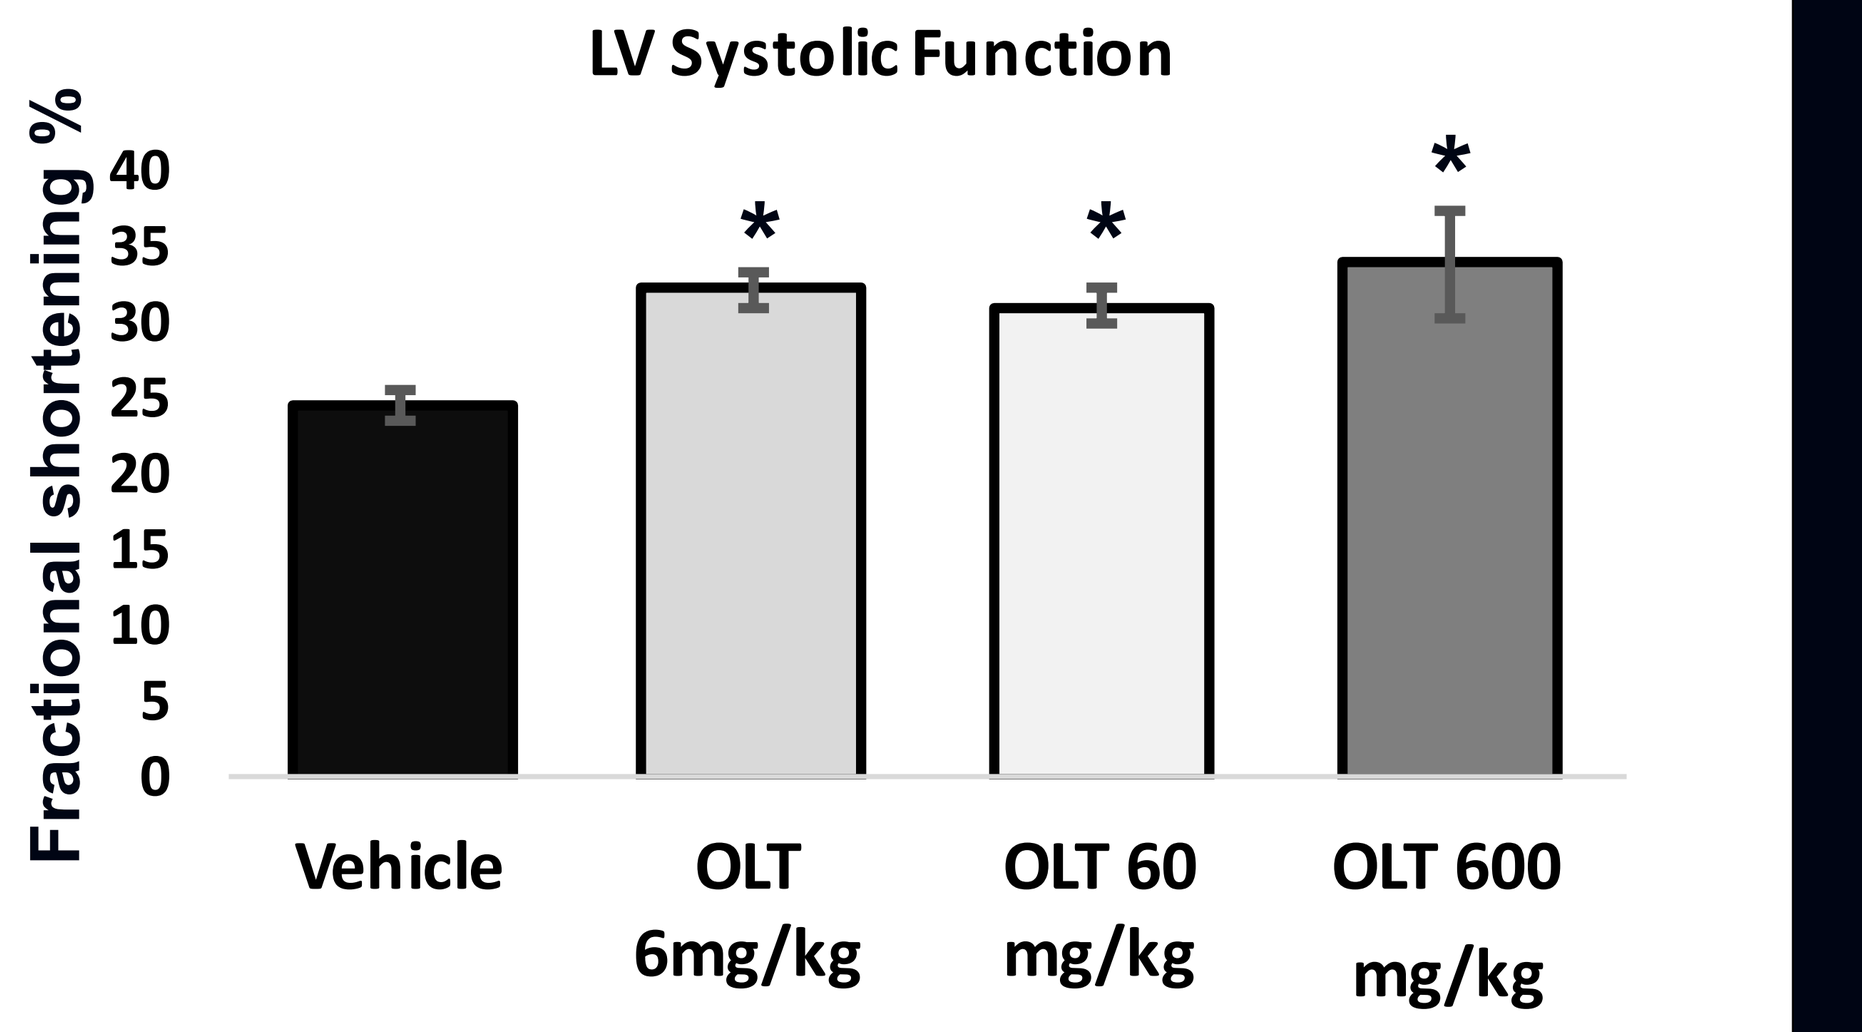
LV Systolic Function
Fractional shortening %
40
35
30
25
20
15
10
5
0
*
*
*
Vehicle
OLT 6mg/kg
OLT 60 mg/kg
OLT 600 mg/kg

Dose dependent effects of OLT1177. OLT1177 (6, 60, or 600 mg/kg) was given at the moment of reperfusion. LVFS and infarct size were measured after 24 hours. N= 5-8 per group. * p<0.005 vs vehicle. ^ p≤0.05 vs OLT 6 mg/kg.

OLT1177 given with 60 minutes delay after reperfusion was equally effective as the treatment given with no delay. After 120 minutes treatment delay, LV systolic function was preserved and infarct size was smaller than in vehicle-treated mice, although the difference was not statistically significant (p=0.052). After 180 minutes treatment delay, OLT1177 failed to prevent the drop in fractional shortening or to reduce the infarct size.

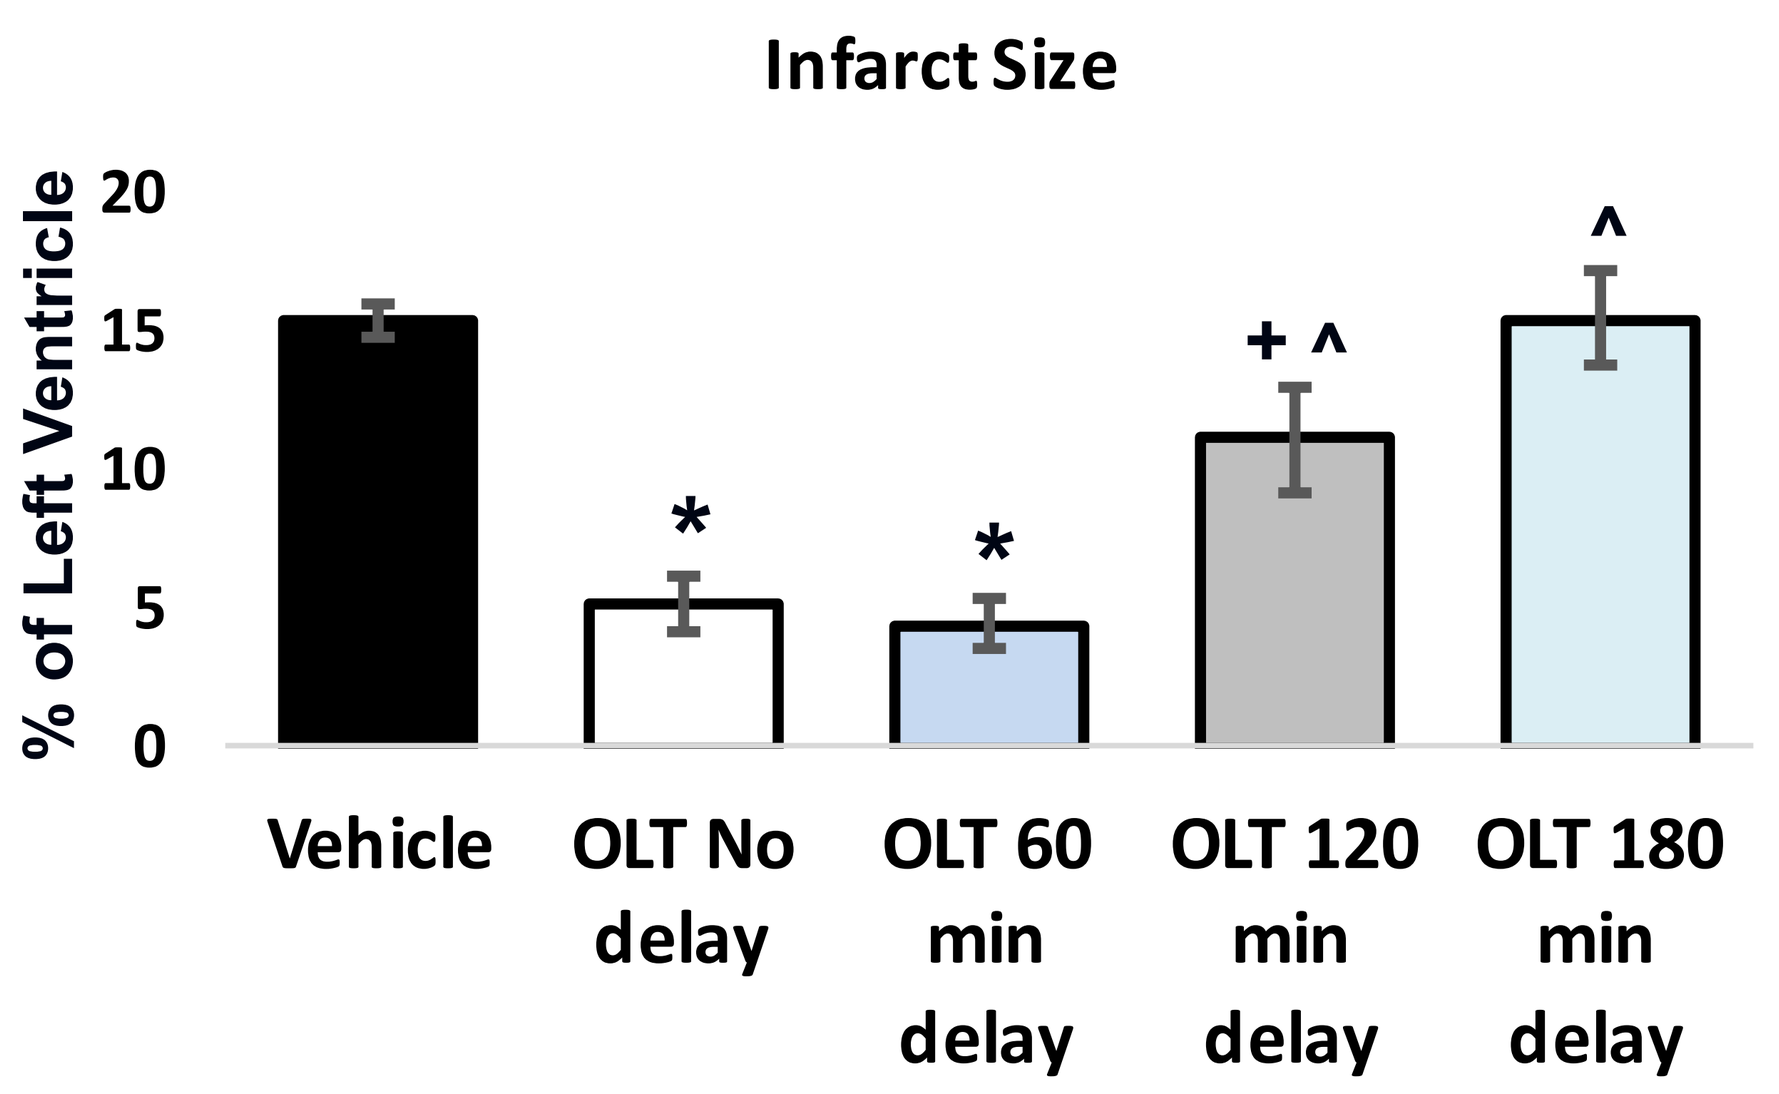
Infarct Size
% of Left Ventricle
20
15
10
5
0
*
*
+ ^
^
Vehicle
OLT No delay
OLT 60 min delay
OLT 120 min delay
OLT 180 min delay

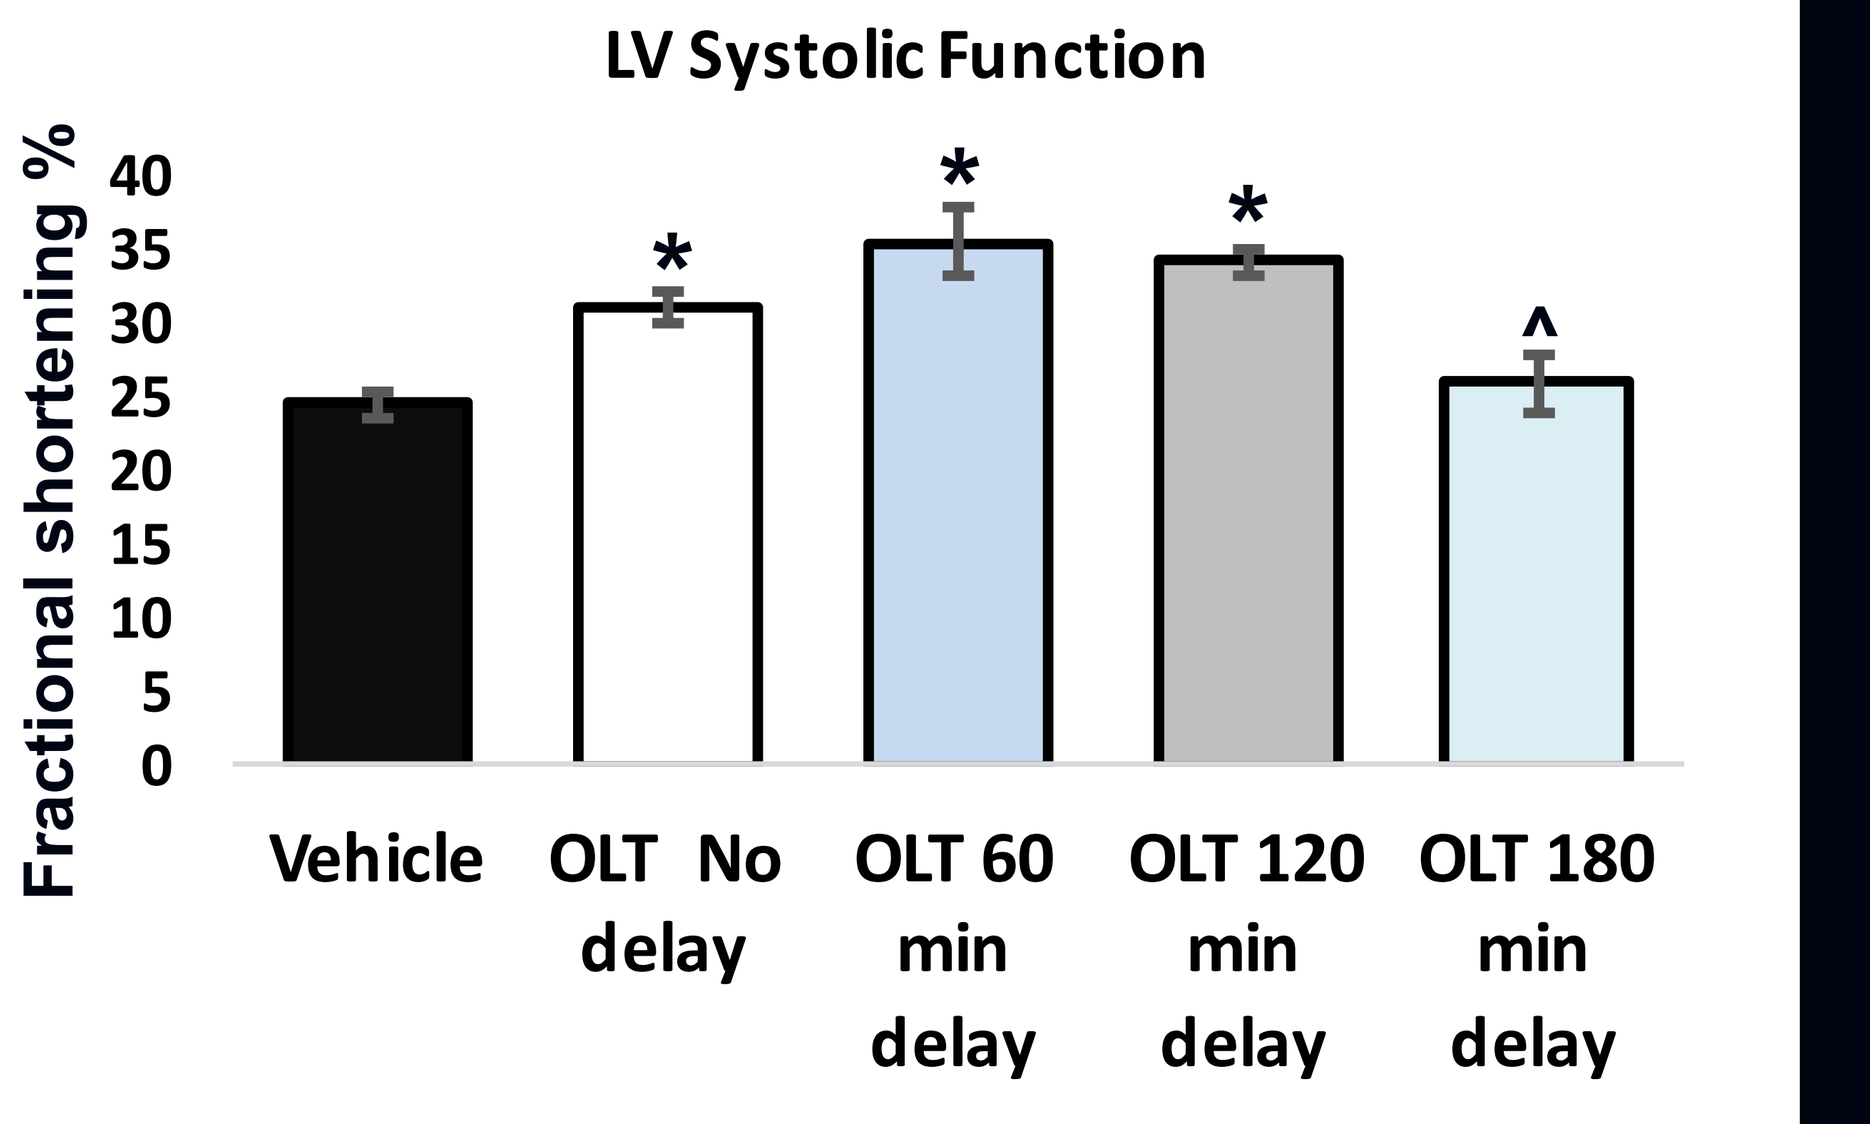
LV Systolic Function
Fractional shortening %
40
35
30
25
20
15
10
5
0
*
*
*
^
Vehicle
OLT No delay
OLT 60 min delay
OLT 120 min delay
OLT 180 min delay

Therapeutic window for delayed OLT1177 treatment. The effects of OLT1177 (60 mg/kg) at intervals of 0, 60, 120, or 180 minutes after reperfusion were compared to the vehicle control. *p<0.005 vs vehicle; ^p<0.05 vs no delay and 60 min delay; ⁺p=0.052 vs vehicle. N=4-7 per group.

Conclusions

Inhibition of the NLRP3 inflammasome using OLT1177, a novel β-sulfonyl nitrile synthetic small molecule, reduces infarct size and preserves LV function in a mouse model of myocardial ischemia/reperfusion injury. These findings provide proof of concept for a protective effect in patients with acute myocardial infarction.

Address for correspondence: Stefano Toldo , PhD, Virginia Commonwealth University, Richmond, VA
stefano.toldo@vcuhealth.org

V i r g i n i a C o m m o n w e a l t h U n i v e r s i t y